New model for quantification of the beat-to-beat instability of the QT duration in drug-induced long QT in anesthetized dogs

A. Van de Water, H. van der Linde, W. Loots, B. Van Deuren, M. Peeters[1], A. Teisman and D. Gallacher.

Center of Excellence for Cardiovascular Safety Research, Johnson & Johnson Pharmaceutical Research & Development

A Division of Janssen Pharmaceutica N.V., B-2340 Beerse, Belgium

[1] XiTechniX, Kleinhoefstraat 5, B-2440 Geel, Belgium.

Introduction

In recent years, several non-cardiovascular drugs have been withdrawn from the market due to cardiovascular side effects. The effects observed were QT interval prolongation and ventricular tachy-arrhythmia known as torsades de pointes (TdP). Since it is recognised that QT interval itself is possibly not the best predictor of risk, the evaluation of better or additional markers is warranted. A more detailed evaluation of the electrophysiological events that underlie TdP would be useful for the identification of torsadogenic drugs in early preclinical development. Markers, among others, associated with congenital and drug-induced TdP include:

(1) prolongation of the ventricular action potential;

(2) generation of early afterdepolarizations (EADs; Tan et.al., 1995);

(3) increased transmural dispersion of ventricular repolarization (TDR; Antzelevitch et al., 2002);

(4) increased temporal dispersion of ventricular repolarization (instability; Hondeghem et al., 2001).

In the Poincaré plot each beat is plotted against the next. Thus it provides a summary of detailed beat-to-beat information, with simple visual interpretation of the data. Currently, adequate quantitative measures that characterize the features of the plot are lacking (Brennan et al., 2001).

In this study:

(1) We provide a novel mathematical model for quantification of the beat-to-beat analysis (i.e. Poincaré plot) of the QT interval of the ECG.

(2) We link these parameters to the morphological properties of the plot; the width being a measurement for the short-term instability (STI), the length for the long-term instability (LTI) and a width and length dependent parameter as an indication for the total instability (TI).

(3) The newly developed parameters are challenged with an established pharmacological compound, dofetilide (class III antiarrhythmic agent; I_{Kr} blocker), to mimic drug-induced long QT.

Methods

Female, adult beagle dogs were used in this study, their body weight averaging 12.0 kg (range: 10.0-13.3 kg). Under total intravenous anesthesia (lofentanil, scopolamine, succinylcholine and infusion of etomidate) the dogs were ventilated with 30% oxygen in pressurized air to normocapnia. The electrocardiogram (ECG lead II limb leads) and the monophasic action potential signal (MAP; bi-directional steerable diagnostic MAP catheter, placed at the endocardium of the right ventricle) were continuously monitored.

Standard cardio-respiratory measurements were taken from each animal but for the sake of clarity only the ECG and MAP variables are discussed. Calculation of the instability parameters was performed before and after administration of solvent (n=7) or dofetilide (n=7) in a period where prolongation of the QT interval reached a steady state (5 min after administration). In the same periods, QT intervals (QT), QT intervals corrected for changes in heart rate according to Van de Water, RR intervals (RR) and monophasic action potential duration to 90% repolarization ($MAPD_{90}$) were automatically analyzed (Notocord-Hem 3.3 software) and reproduced as median values over 1 minute. An EAD was defined as a small afterpotential that interrupts or delays the normal repolarization of the action potential.

After at least 20 min recording stable signals, the compound or solvent solutions were slowly (± 10 seconds) injected intravenously with increasing doses every 30 minutes. Solvent was administered in equivalent volumes with the compound (sodium chloride 0.9%; volumes from 0.025 to 0.4 ml/kg i.v.; total volume = 1.97 ml/kg i.v.). Dofetilide (solution of 0.1 mg/ml in saline) was administered in a dose range of 0.0025, 0.005, 0.01, 0.02 and 0.04 mg/kg i.v.. Arterial blood samples for measurement of dofetilide plasma concentrations were taken at 5 and 30 minutes after each injection of dofetilide in 4 experiments. The heparinised blood was centrifuged within 10 min and the plasma stored at -20°C until analysis by LC-MS/MS with a limit of quantification of 1 ng/ml.

Before performing the quantitative analysis of the QT-instability, visual examination of the ECG tracings was done. Special emphasis was placed on artifact detection and elimination, because these would strongly affect instability analysis of the ECG signal. Starting from a total trend graph of all QT intervals (Figure 1), an ECG tracing of 30 qualified consecutive beats (no artifacts or ectopic beats) was chosen for further analysis. All these tracings were visualized by Poincaré plots (Figure 2).

Mathematical model

Figure 1: A trend graph of beat-to-beat QT intervals of one individual experiment is represented below. Dofetilide was administered at increasing doses of 0.0025 to 0.04 mg/kg at 30 min intervals. The moment of the administration is pointed with an arrow. The dotted lines indicate the places were the 30 beat tracings are picked. The width of the line of the QT intervals is a rough indication for the QT instability.

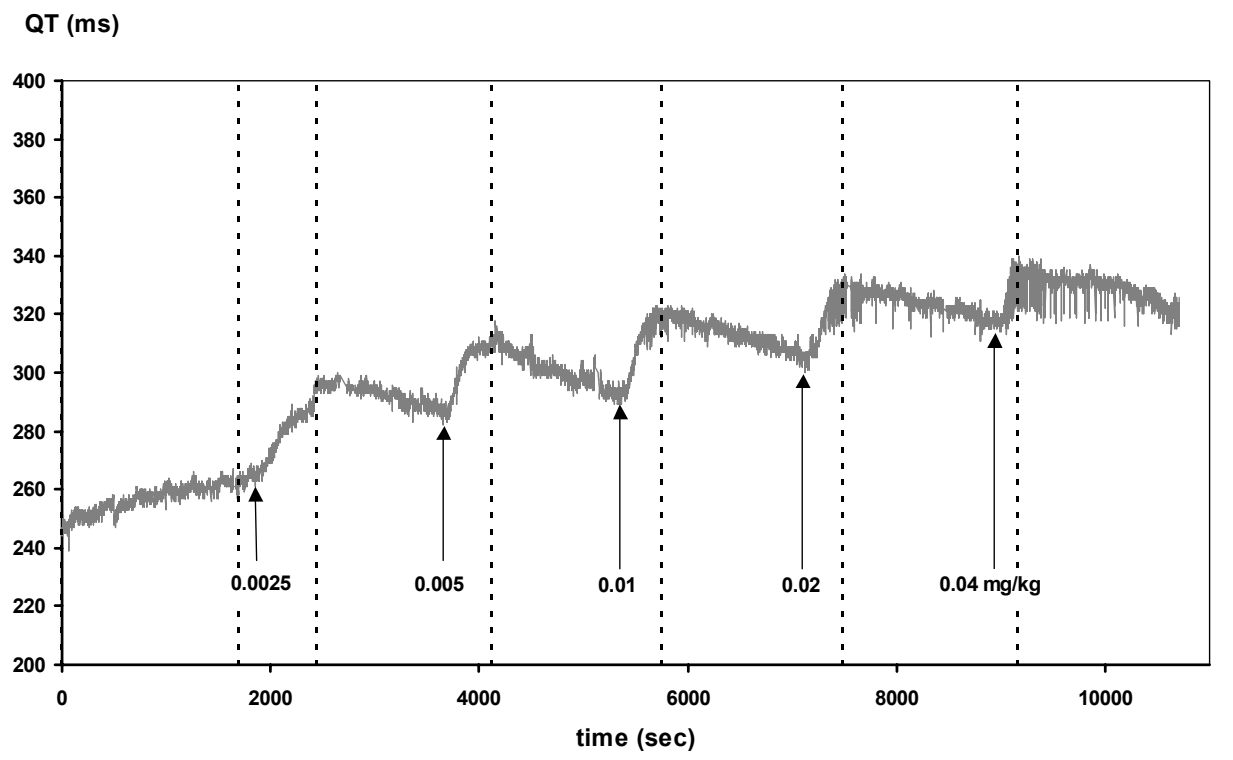
QT (ms)
400
380
360
340
320
300
280
260
240
220
200
0.0025
0.005
0.01
0.02
0.04 mg/kg
0
2000
4000
6000
8000
10000
time (sec)

Figure 2: Selected tracings are automatically visualized in Poincaré plots and manually checked for artifacts or ectopic beats. This example shows the dose-dependent effect of increasing doses of dofetilide in one anesthetized dog.

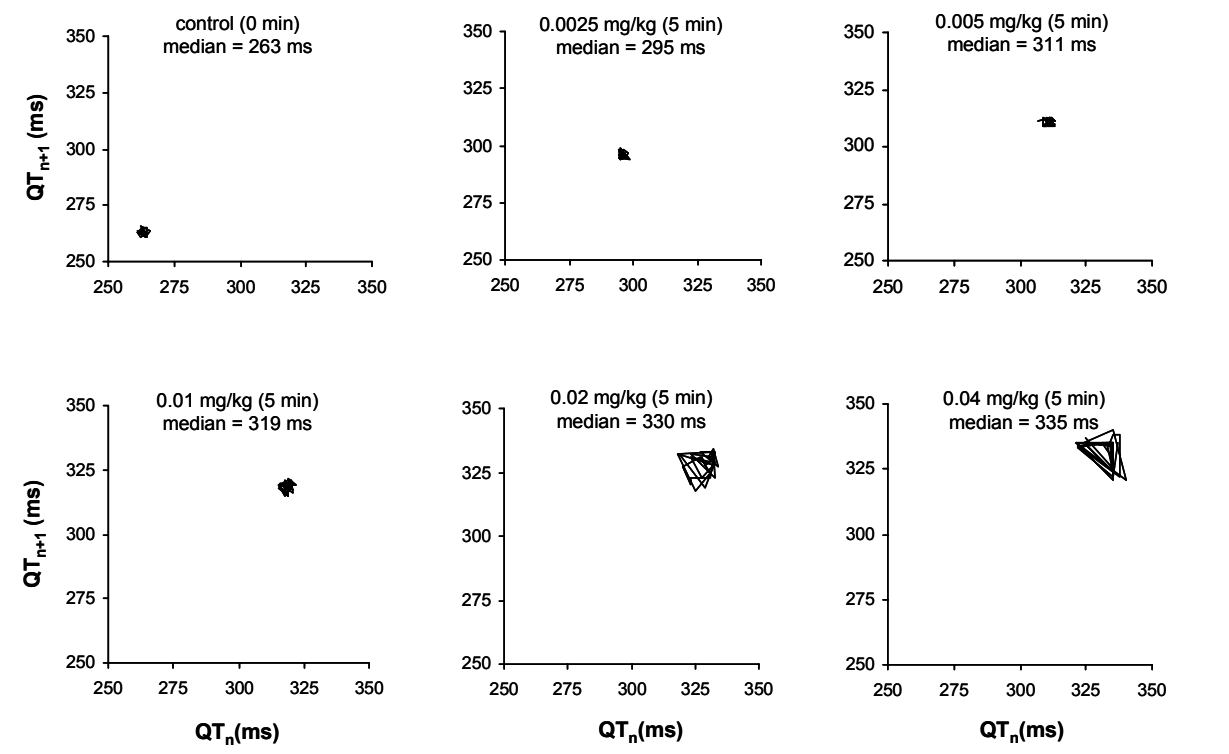
control (0 min)
median = 263 ms
0.0025 mg/kg (5 min)
median = 295 ms
0.005 mg/kg (5 min)
median = 311 ms
0.01 mg/kg (5 min)
median = 319 ms
0.02 mg/kg (5 min)
median = 330 ms
0.04 mg/kg (5 min)
median = 335 ms
QTn+1 (ms)
QTn(ms)
250
275
300
325
350

Figure 3: Quantification of the Poincaré plot is based on the distances to the center of gravity of a cluster of 30 QT intervals. Only 28 data points are identifiable because 2 points are exactly the same. That's why visual examination is not always accurate. Notice that in this example the center of gravity is not exactly located on the line of identity.

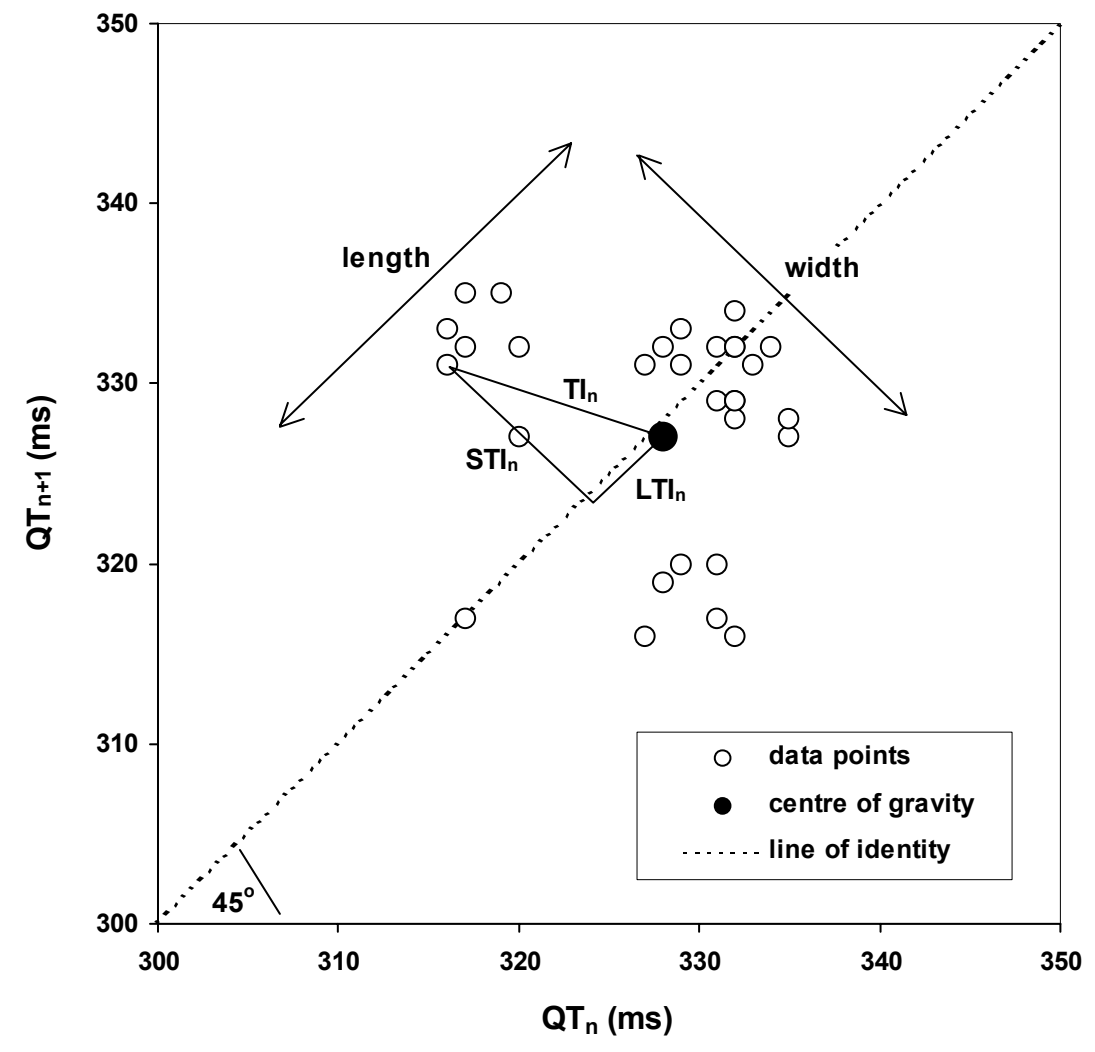
length
width
TIn
STIn
LTIn
45°
data points
centre of gravity
line of identity
QTn+1 (ms)
QTn (ms)
300
310
320
330
340
350

To quantify the plot we first calculated the center of gravity (cg) of the cluster of data points according to following equation:

center of gravity (cg): $$cg(x) = \sum_{i=m}^{m+29} (QT_i) \Big/ 30 \qquad cg(y) = \sum_{i=m+1}^{m+30} (QT_i) \Big/ 30$$

Afterwards, the distances of all 30 data points to the cg were calculated. The median of these distances is the total instability (TI) over the 30 points (Figure3):

Total Instability: $$TI_n = \sqrt{\left((cg(x) - QT_n)^2 + (cg(y) - QT_{n+1})^2\right)}$$

$$TI = M(TI_n)$$

To measure the length and the width of the plot we performed a rotation of -45° around the origin and obtained following mathematical formulae:

Rotated cg(x): $$Rcg(x) = (\cos\theta \times cg(x)) - (\sin\theta \times cg(y))$$

Rotated cg(y): $$Rcg(y) = (\sin\theta \times cg(x)) + (\cos\theta \times cg(y))$$

In the reference system of the new [Rcg(x),Rcg(y)], coordinates of all distances were measured to the x coordinate of the center of gravity and to the y coordinate of the center of gravity. The distance to the x coordinate represents the length of the plot and reflects the long-term instability (LTI) and the distance to the y coordinate is the width of the plot and reflects the short-term instability (STI). After calculation of these distances for all data points, the median value is calculated for the instability parameters of the total tracing of 30 beats.

Long-Term Instability: $$LTI_n = \left|Rcg(x) - ((\cos\theta \times QT_{n+1}) - (\sin\theta \times QT_n))\right|$$

$$LTI = M(LTI_n)$$

Short-Term Instability: $$STI_n = \left|Rcg(y) - ((\sin\theta \times QT_{n+1}) + (\cos\theta \times QT_n))\right|$$

$$STI = M(STI_n)$$

These three instability parameters (TI, LTI and STI) were measured for all individual dogs.

Glossary:

n	number of the beat from 1 to 29
m	the first beat of a selected tracing
cg(x)	x-coordinate of centre of gravity
cg(y)	y-coordinate of centre of gravity
M	median over 30 beats
θ	$-\pi/4$

Results

None of the measured and calculated parameters were influenced by the solvent treatments. Dofetilide dose-dependently increased QT, QTc and QT Instability. Maximal QT and QTc prolongation was 12 % *vs* baseline at 0.01 mg/kg i.v. (Mean plasma concentration: C_{mean} = 11 ng/ml). TI and STI achieved their maximum at 0.04 mg/kg i.v. (C_{mean} = 40 ng/ml; 434 % and 481 % *vs* baseline, respectively) and LTI at 0.02 mg/kg i.v. (C_{mean} = 18 ng/ml; 598 % *vs* baseline). EADs developed after low doses of dofetilide (0.005 and 0.01 mg/kg i.v.) in 43% of the experiments (3 out of 7 dogs); at higher doses (0.02 and 0.04 mg/kg i.v.) EADs developed in 86% of the animals (6 out of the 7 dogs) (Table 1).

Table 1: Dose-dependent changes of QT, QTc and QT Instability and occurrence of EADs after administration of various doses of dofetilide in anesthetized dogs.

		Dofetilide (n=7)				
	baseline	0.0025 (mg/kg)	0.005 (mg/kg)	0.01 (mg/kg)	0.02 (mg/kg)	0.04 (mg/kg)
QT (ms)	250 ± 6	266 ± 6†	277 ± 8*†	279 ± 11*†	279 ± 13*†	280 ± 15*†
QTc (ms)	260 ± 3	272 ± 3	284 ± 7*†	290 ± 9*†	289 ± 10*†	287 ± 9*†
TI (ms)	1.4 ± 0.1	1.8 ± 0.1	2.8 ± 0.5	3.3 ± 0.5†	5.2 ± 1.1*†	6.8 ± 0.9*†
STI (ms)	0.7 ± 0.0	1.0 ± 0.2	1.4 ± 0.3	1.9 ± 0.3†	2.4 ± 0.3*†	4.2 ± 0.6*†
LTI (ms)	0.7 ± 0.1	0.7 ± 0.0	1.6 ± 0.3	2.0 ± 0.3	3.7 ± 1.2*	3.6 ± 0.5*†
EAD (n / 7)	0 / 7	1 / 7	3 / 7	3 / 7	6 / 7 #	6 / 7 #

QTc: QT corrected according to Van de Water, TI: Total Instability, STI: Short-Term Instability, LTI: Long-Term Instability, EAD: early afterdepolarization. All values are mean ± sem.

* = $p < 0.05$ (analysis based on a repeated measurements ANOVA model followed by a Newman-Keuls multiple comparisons test: effect after dose compared to baseline)

† = $p < 0.05$ (analysis based on a Dunnett's test: comparison between solvent and dofetilide treatment).

\# = $p < 0.05$ (Fisher's probability test was used to evaluate the differences in the incidence of EADs between the two groups).

Conclusion

Measuring length and width of Poincaré plots, by calculating the distances to the y- and x-coordinate of the center of gravity to the data points is a valuable measurement for long-term and short-term instability.

Total instability is introduced as the median distance of the data points to the center of gravity, and is a parameter build up by the length (long-term) and the width (short-term) of the Poincaré plot.

Significant QTc prolongation in this model is identified at clinically relevant doses and exposures. Indeed, inhibition of I_{Kr} channels in anesthetized dogs with dofetilide also induces EADs in the monophasic action potential and increases QT instability at higher doses.

As such, the proposed quantification parameters of QT Poincaré plots have added value as an additional marker for performing risk assessments using *in vivo* data. Further work is needed to benchmark the potential benefits of this biomarker against other markers.

References

1. Antzelevitch, C., Shimizu, W. (2002). Cellular mechanisms underlying the QT syndrome. *Current Opinion in Cardiology*, 17, 43-51.
2. Brennan, M., Palaniswami, M., Kamen, P. W. (2001). Do existing measures of Poincaré plot geometry reflect nonlinear features of heart rate variability? *IEE Transactions on Biomedical Engineering*, 48, 1342-1347.
3. Hondeghem, L. M., Carlsson, L., Duker, G. (2001). Instability and triangulation of the action potential predict serious proarrhythmia, but action potential duration prolongation is antiarrhythmic. *Circulation*, 103, 2004-2013.
4. Tan, H.L, Hou, C.J.Y, Lauer, M.R., Sung, R.J. (1995). Electrophysiologic mechanisms of the long QT interval syndromes and torsade de pointes. *Annals of Internal Medicine*, 122, 701-714.